Sandwell and West Birmingham Hospitals NHS
NHS Trust

Liraglutide is effective across a range of overweight and obese body mass indices; findings from the Association of British Clinical Diabetologists (ABCD) nationwide liraglutide audit

Ryder REJ,[1] Sen-Gupta P,[1] Thong KY[1]

[1]City Hospital, Birmingham, UK

Introduction

- Treatment of type 2 diabetes with glucagon-like peptide (GLP)-1 receptor agonists has been shown to significantly decrease body weight and body mass index (BMI).[1]
- To examine the effectiveness of GLP-1 receptor agonist therapy across a range of baseline BMI, the current analysis used data from a nationwide audit that assessed the treatment response to liraglutide stratified by pre-treatment BMI.

Methods

- Data used in this analysis was obtained from the ABCD nationwide liraglutide audit. The audit collected anonymised clinical data of patients treated with liraglutide in clinical practice. Diabetes centres across UK were invited to participate.

Figure 1. Patients evaluated in audit and reasons for exclusion.

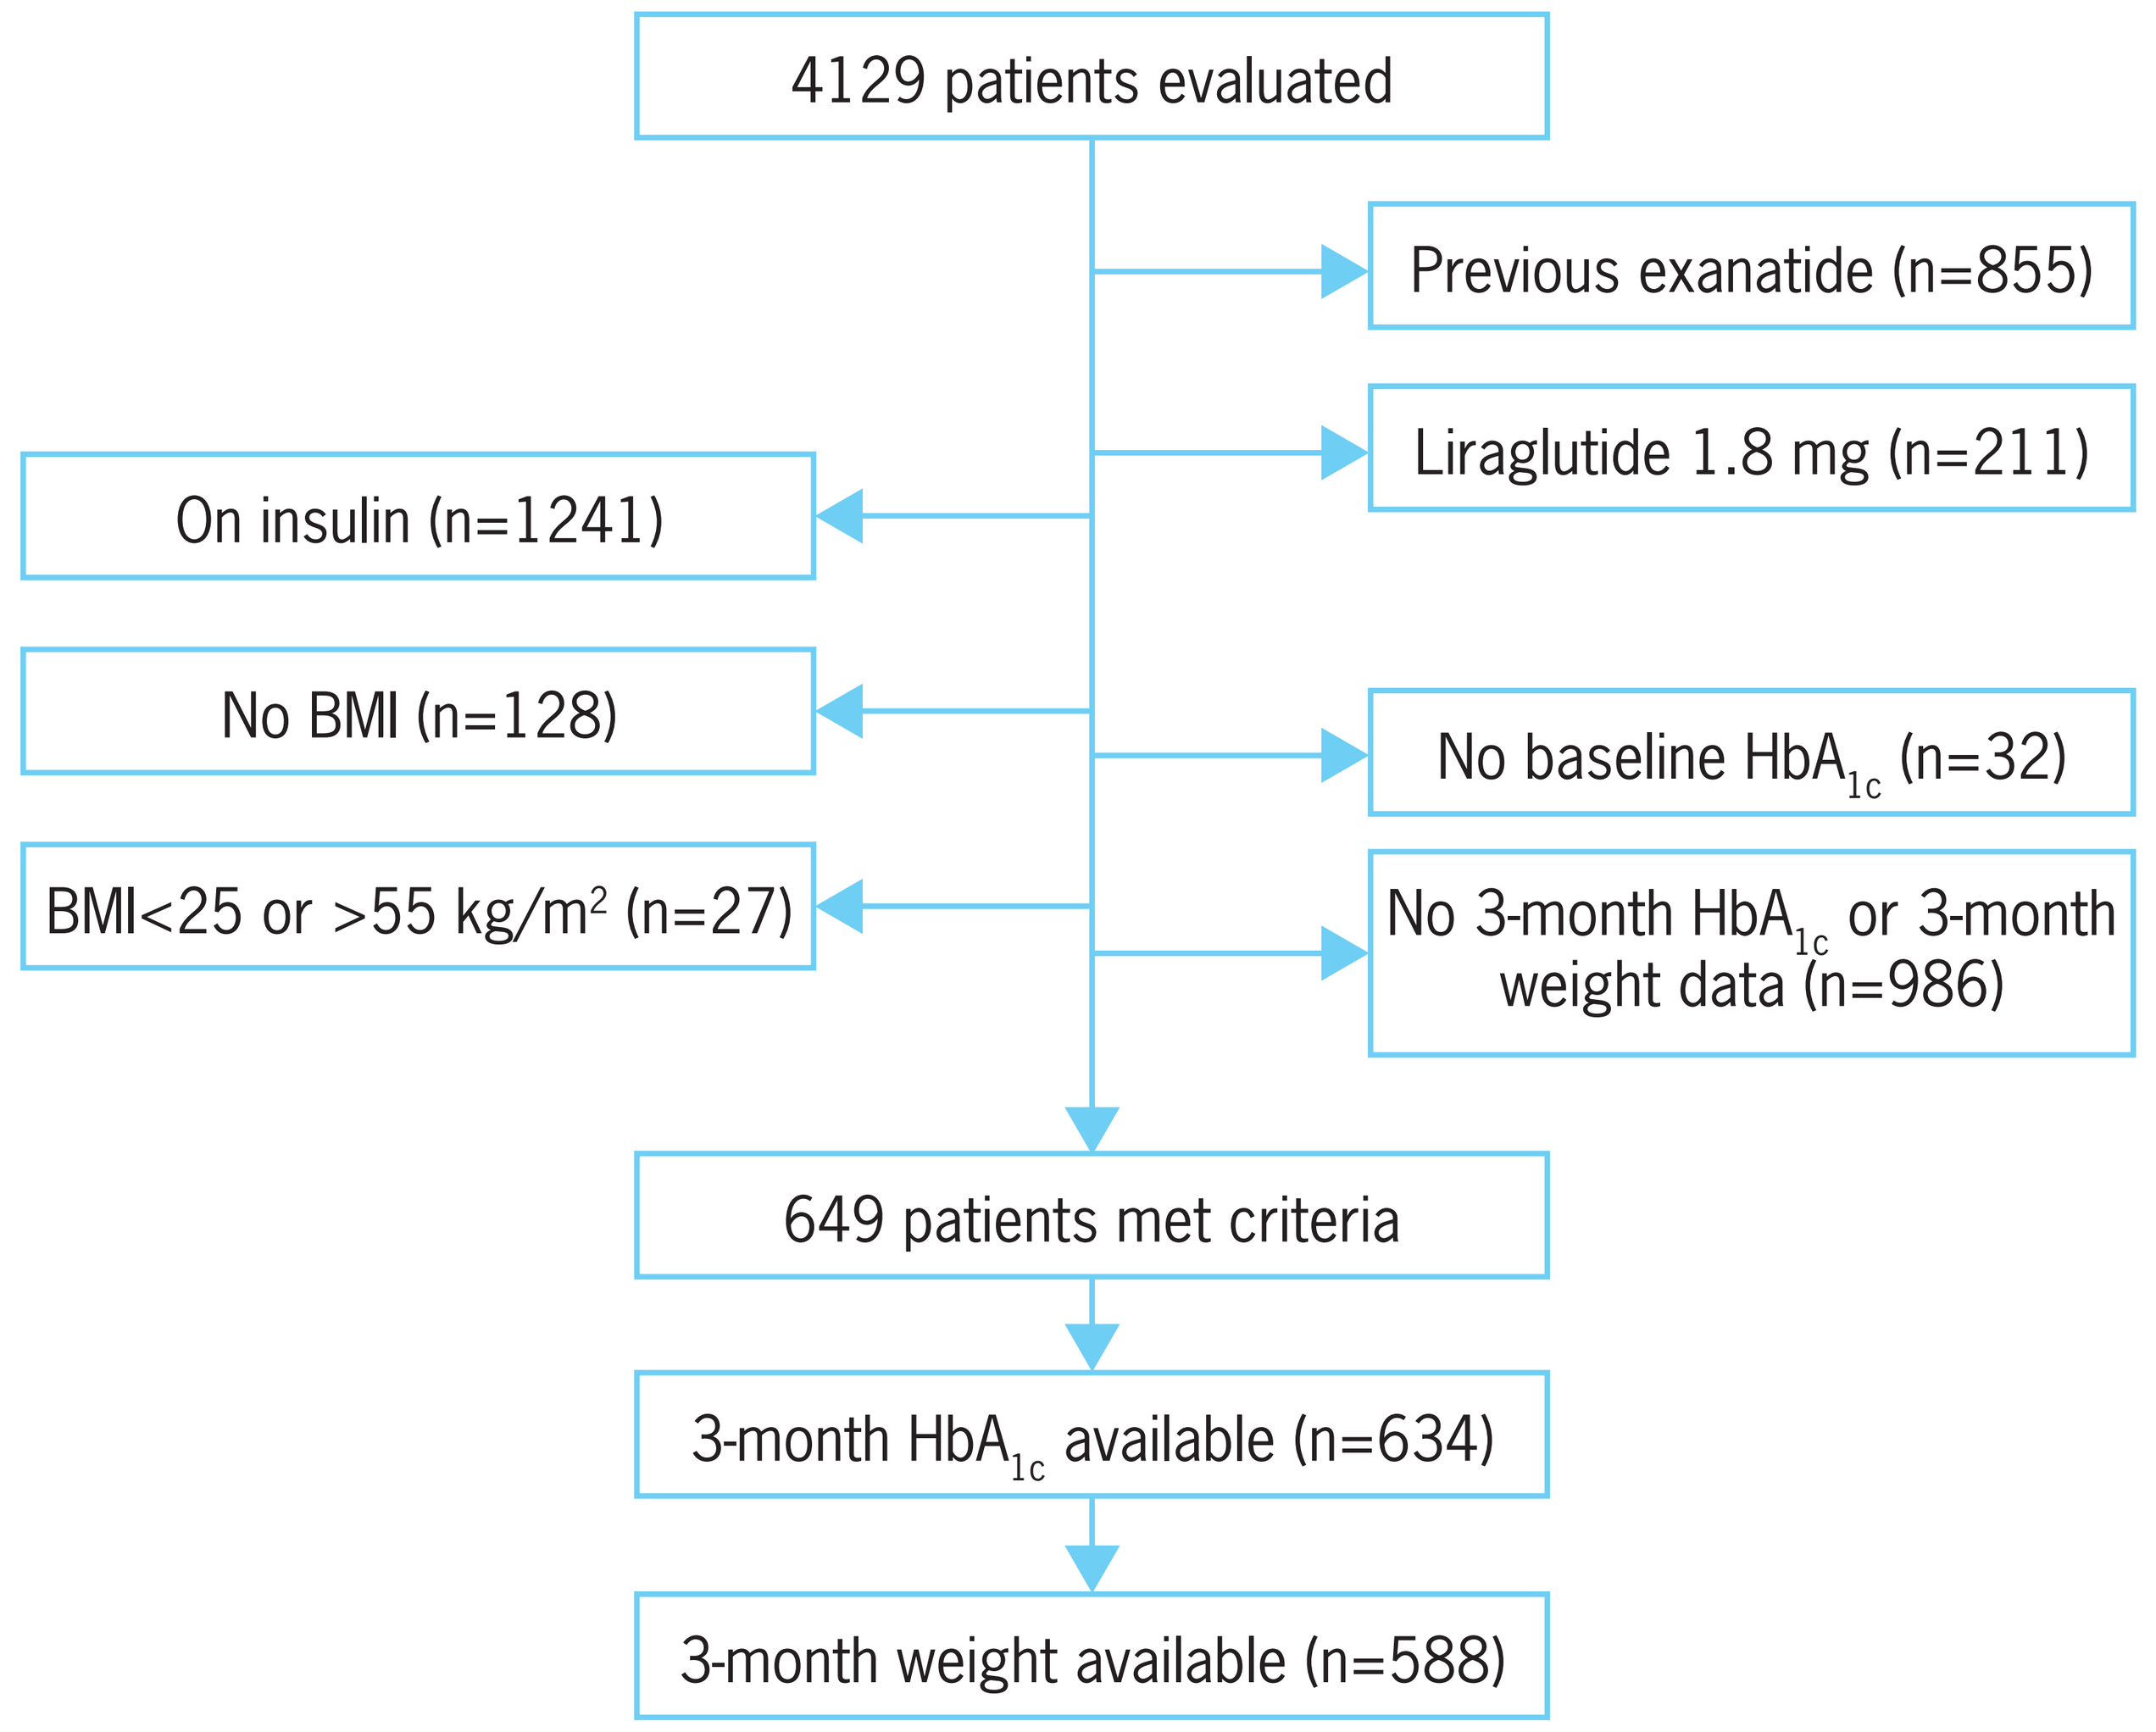
4129 patients evaluated
Previous exanatide (n=855)
Liraglutide 1.8 mg (n=211)
On insulin (n=1241)
No BMI (n=128)
No baseline HbA1c (n=32)
BMI<25 or >55 kg/m2 (n=27)
No 3-month HbA1c or 3-month weight data (n=986)
649 patients met criteria
3-month HbA1c available (n=634)
3-month weight available (n=588)

- Patients who switched from exenatide to liraglutide therapy, were treated with a liraglutide 1.8 mg dose, used insulin, had no BMI data, had BMI <25 kg/m^2 or >55 kg/m^2, were lost to follow-up, or whose follow-up data was outside the specified time frame, were excluded.
- Data on reduction of HbA_{1c}, weight, and weight as a percentage of initial body weight was assessed at 3 months (±6 weeks) of liraglutide treatment.
- Results were compared across six baseline BMI groups: 25–29.9 kg/m^2, 30–34.9 kg/m^2, 35–39.9 kg/m^2, 40–44.9 kg/m^2, 45–49.9 kg/m^2 and 50–54.9 kg/m^2.

Figure 2. Change in HbA_{1c}* by baseline BMI category at 3 months of liraglutide 1.2 mg treatment.

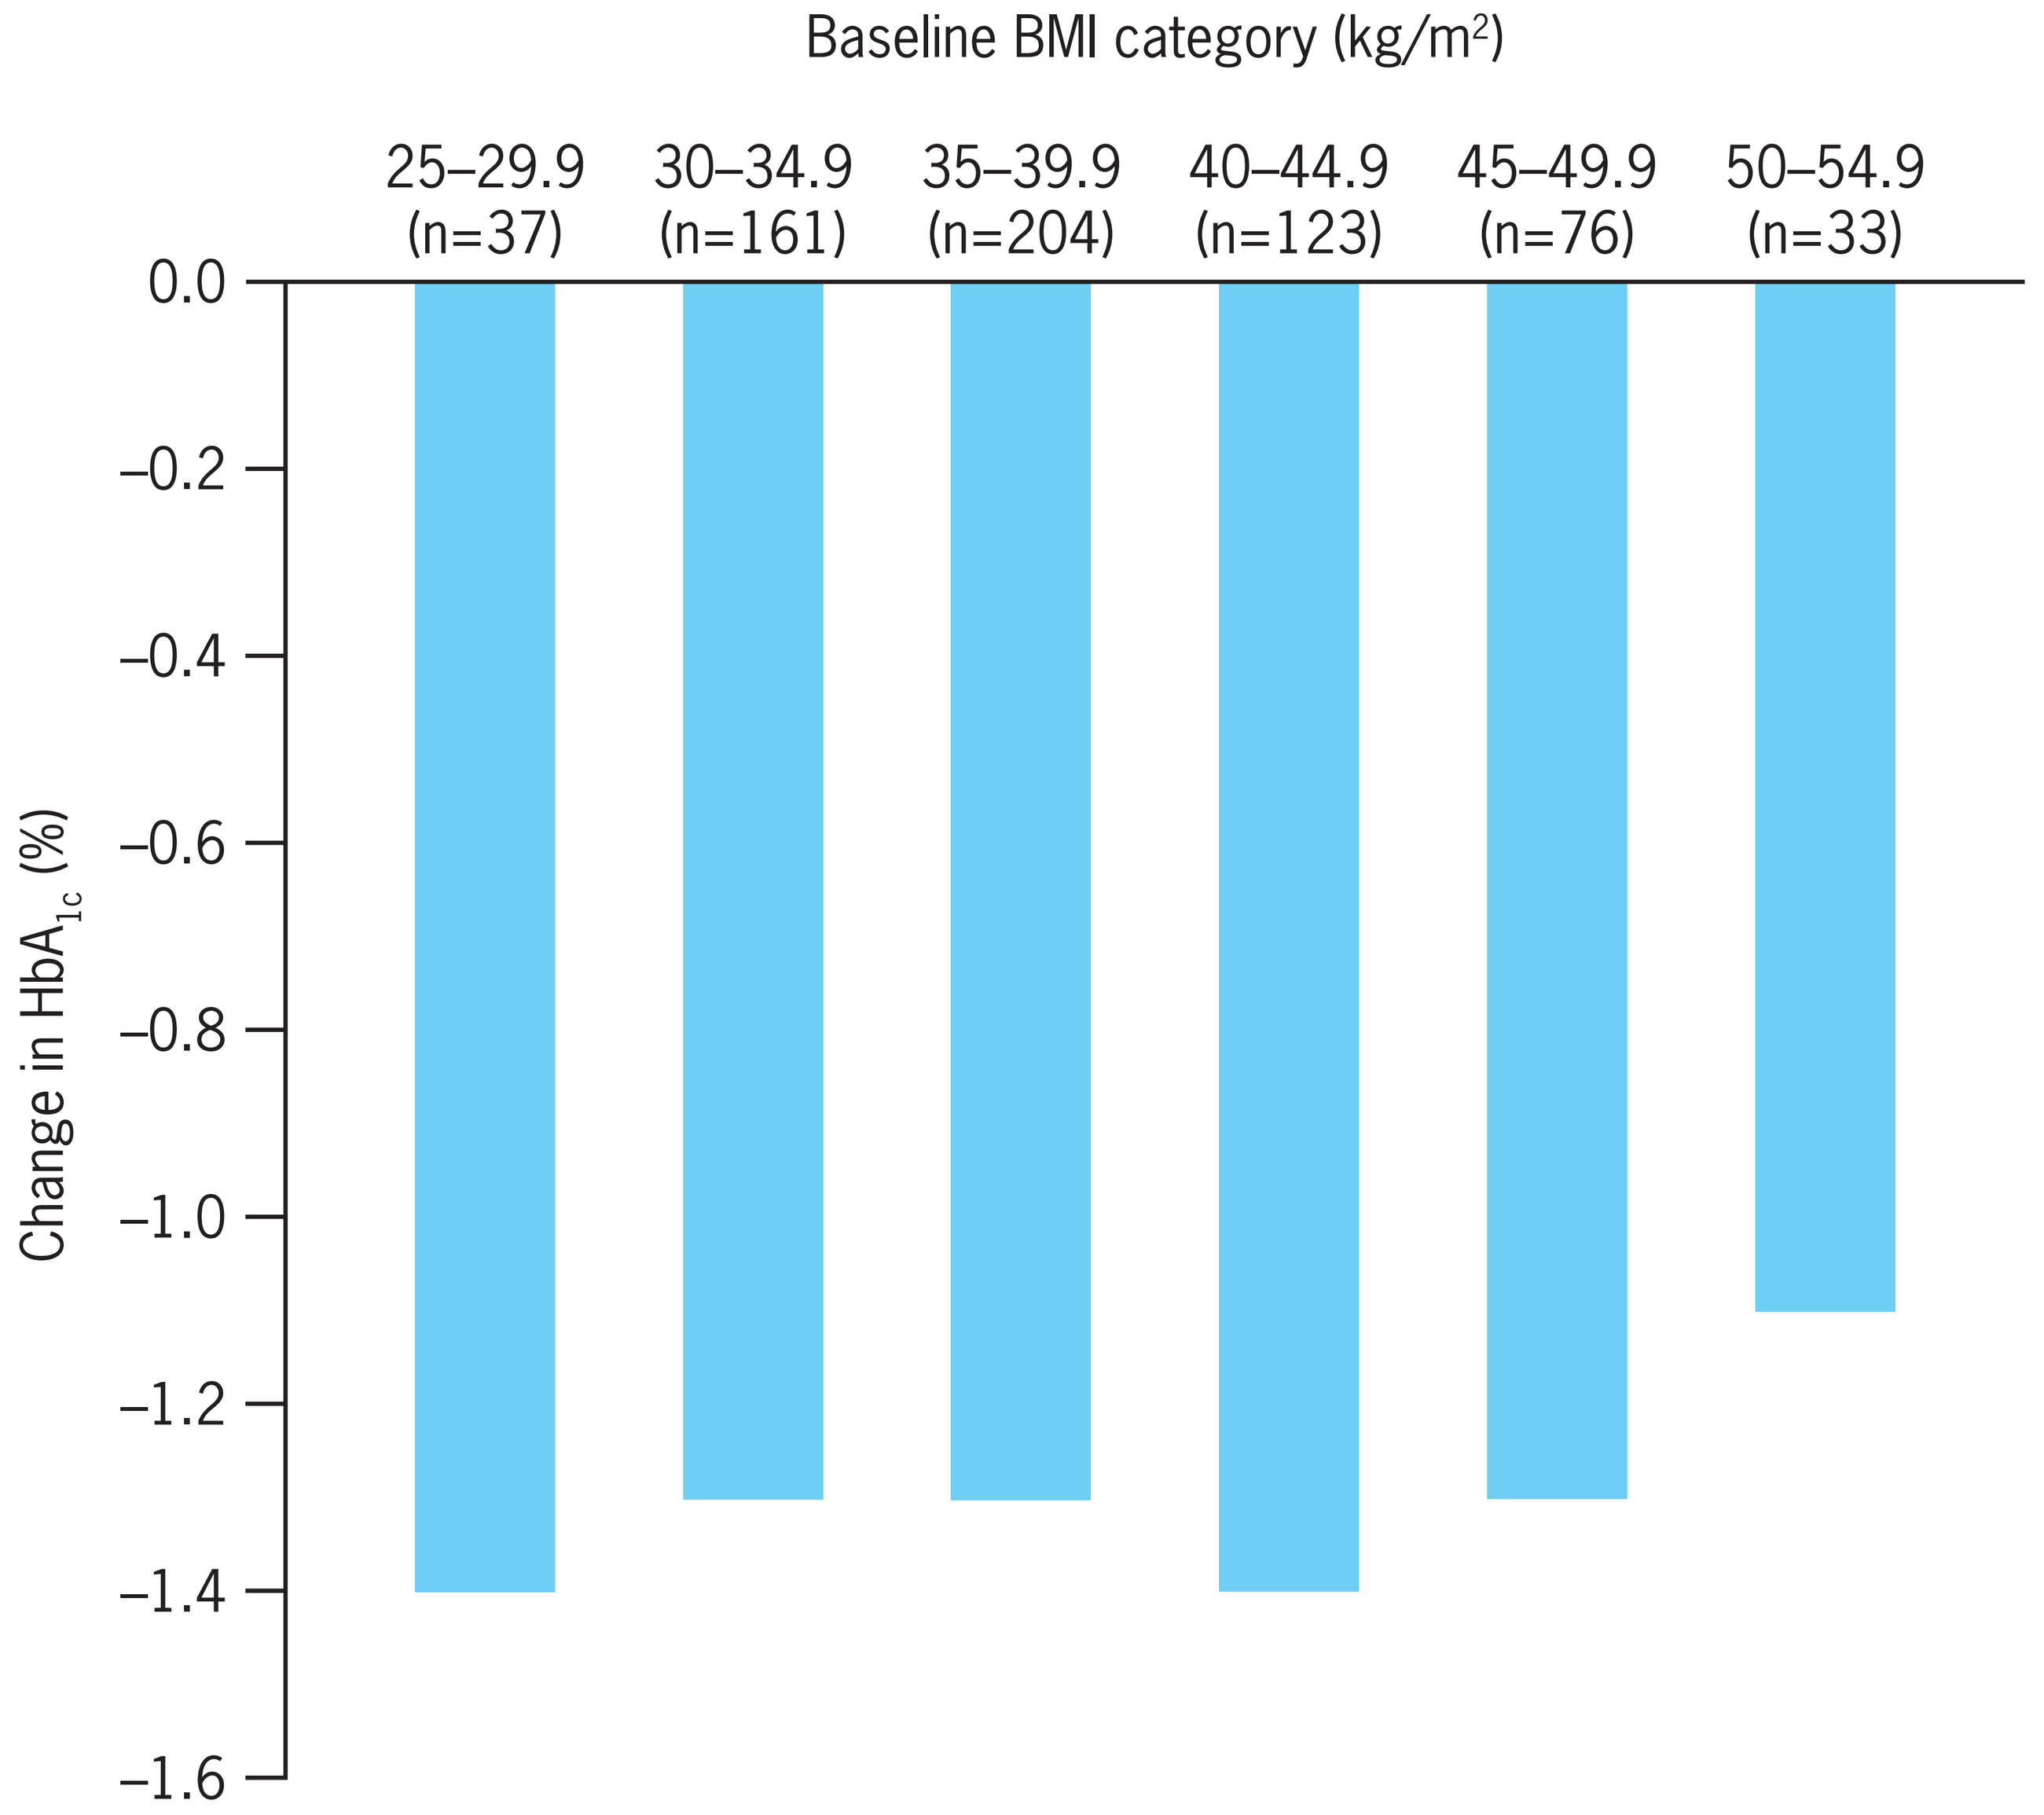
Baseline BMI category (kg/m2)
25–29.9 (n=37)
30–34.9 (n=161)
35–39.9 (n=204)
40–44.9 (n=123)
45–49.9 (n=76)
50–54.9 (n=33)
0.0
–0.2
–0.4
–0.6
–0.8
–1.0
–1.2
–1.4
–1.6
Change in HbA1c (%)

*Mean HbA_{1c} was adjusted for baseline HbA_{1c}, ethnicity, gender, age and number of oral antidiabetes drugs. p=0.894 for effect of baseline BMI.

Results

- Data were collected from 77 centres on 4129 patients with mean (SD) HbA_{1c} of 9.4% (1.7) and BMI of 38.8 kg/m^2 (7.3). After exclusions, there were 634 HbA_{1c} and 588 weight datapoints for comparison among the six BMI groups (Figure 1).
- Mean HbA_{1c} reductions were between 1.1% to 1.4%, and no difference in HbA_{1c} reduction between BMI groups was found either before (p=0.929) or after adjustment for factors such as baseline HbA_{1c} and ethnicity (p=0.894) (Figure 2).

Figure 3. Change in weight (kg) by baseline BMI category at 3 months of liraglutide 1.2 mg treatment.

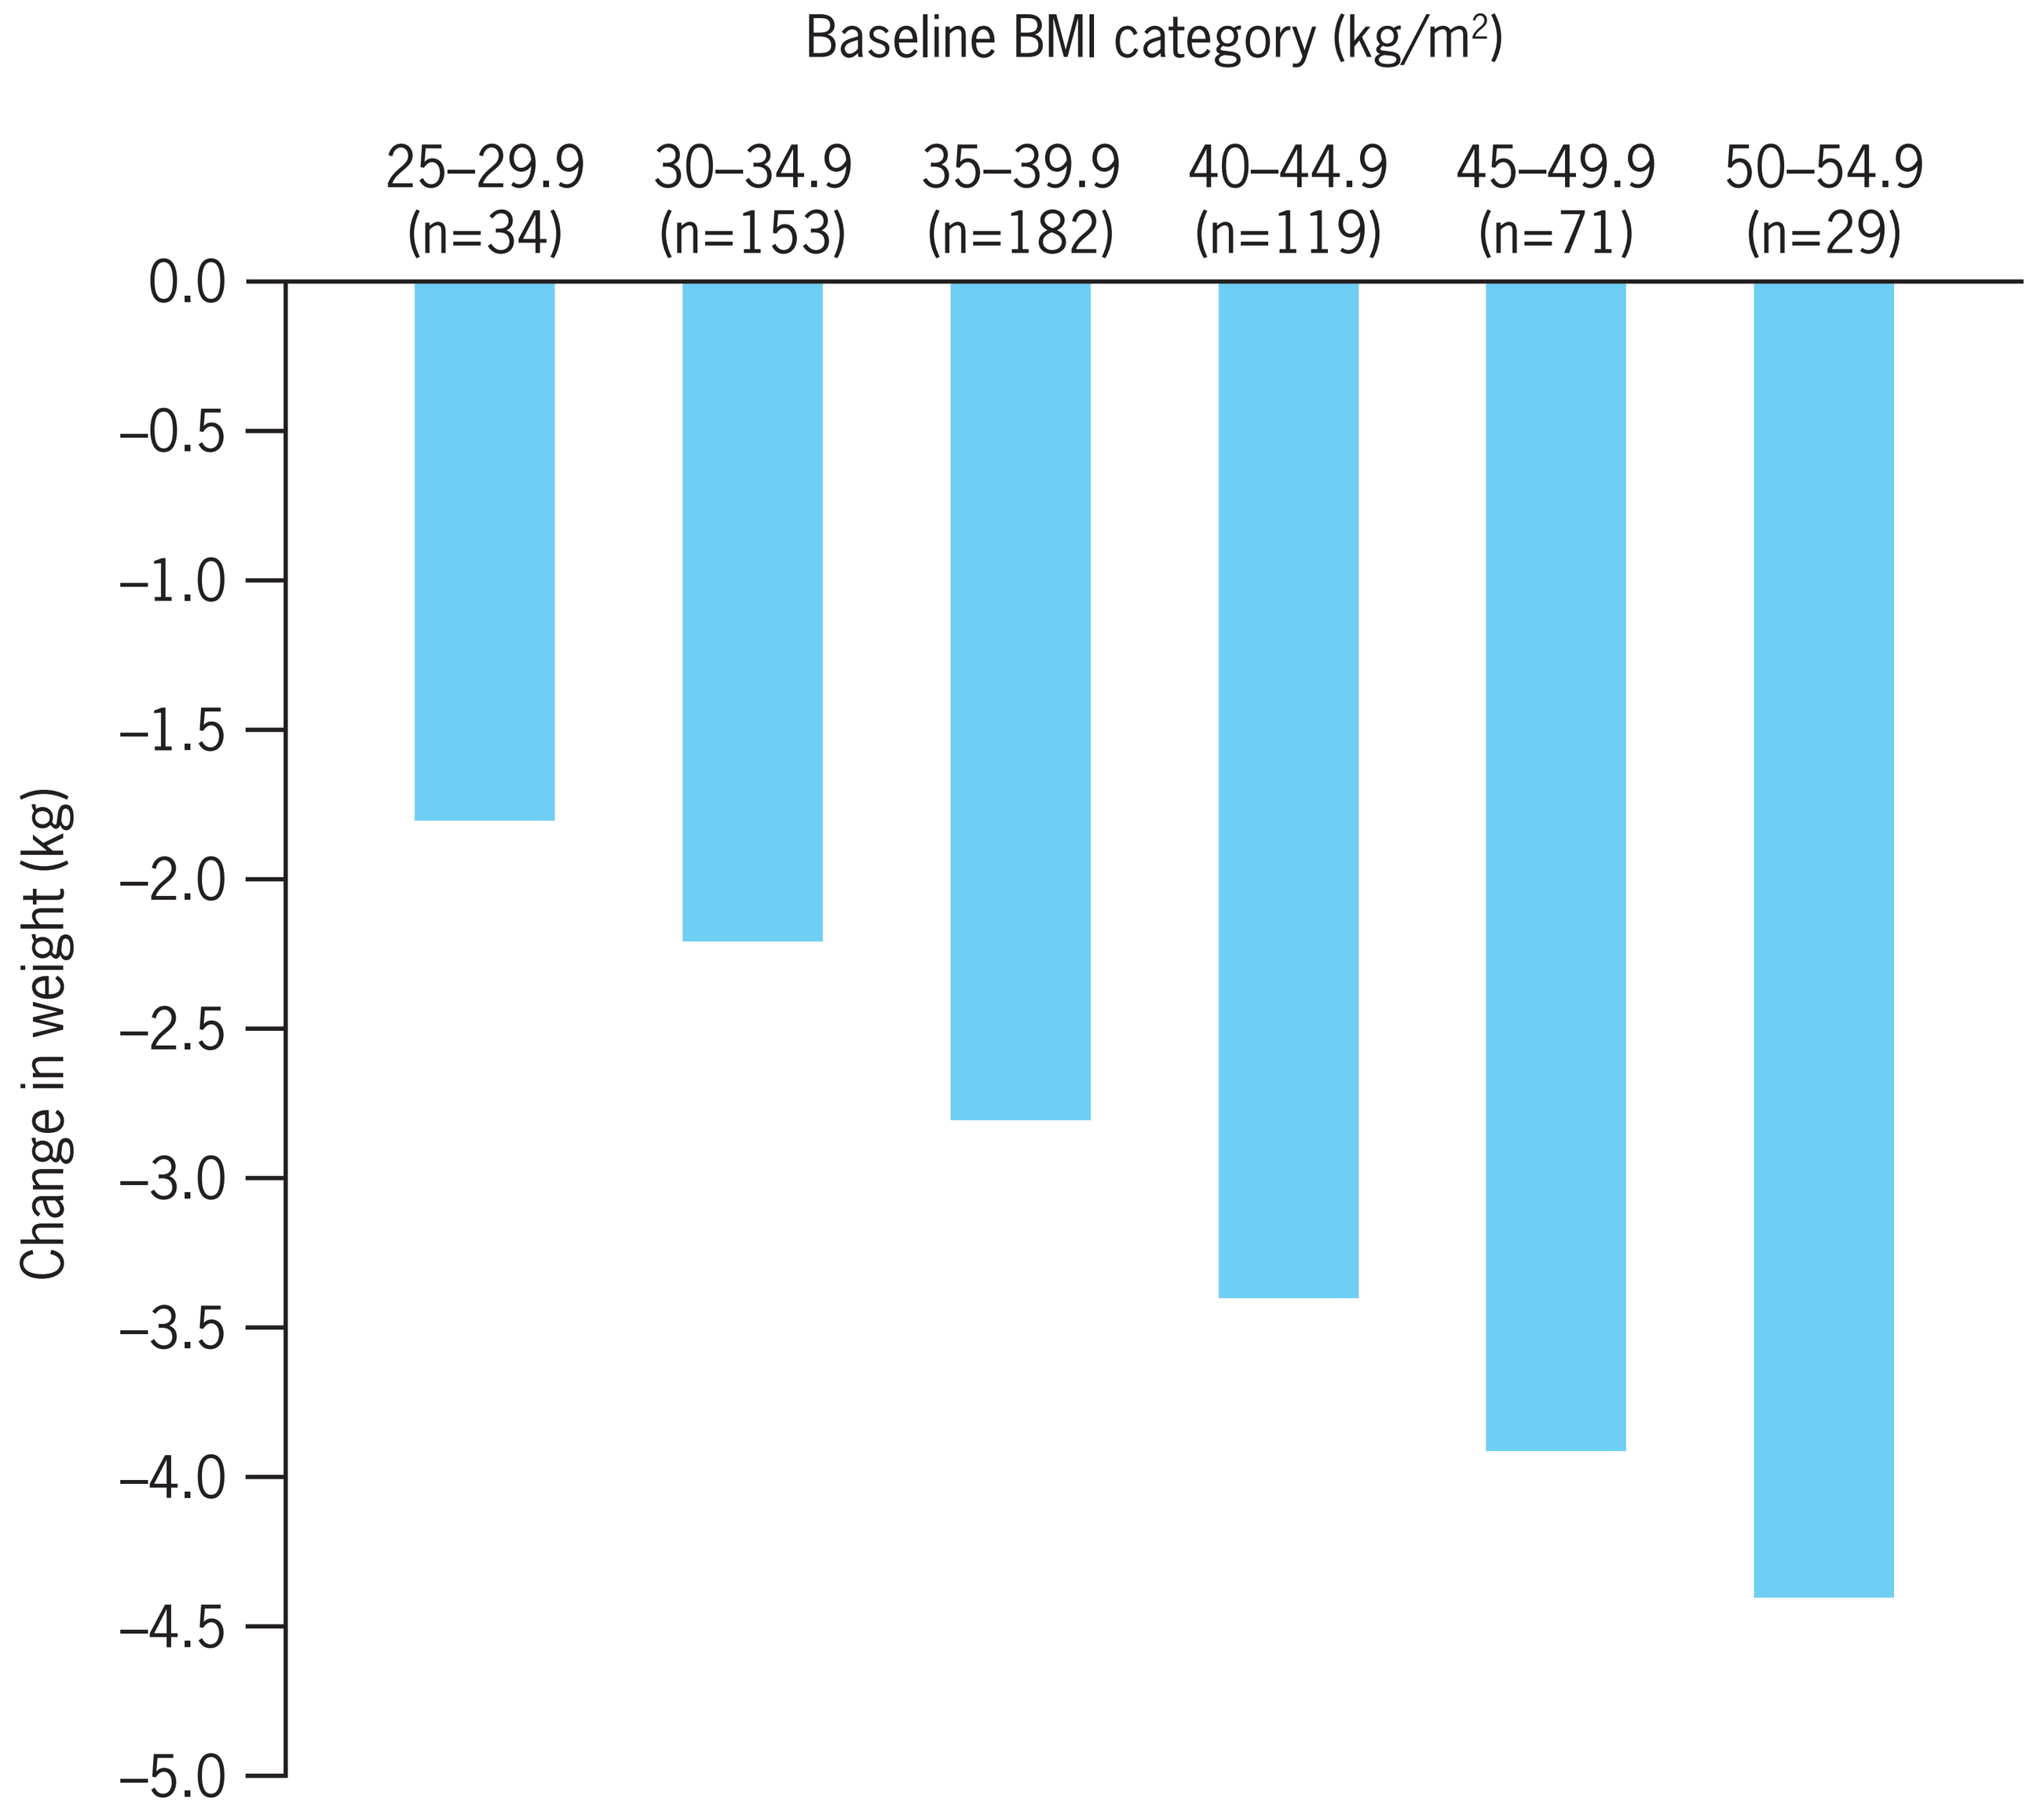
Baseline BMI category (kg/m2)
25–29.9 (n=34)
30–34.9 (n=153)
35–39.9 (n=182)
40–44.9 (n=119)
45–49.9 (n=71)
50–54.9 (n=29)
0.0
–0.5
–1.0
–1.5
–2.0
–2.5
–3.0
–3.5
–4.0
–4.5
–5.0
Change in weight (kg)

Overall p=0.010 for effect of baseline BMI.

- Weight loss, ranging from 1.8 kg to 4.4 kg, increased significantly with greater baseline BMI (p=0.010) (Figure 3). However, weight loss as a percentage of initial body weight was similar across BMI groups (ranging from –2.2% to –3.1% of initial body weight; p=0.649).

Conclusions

- The use of liraglutide in the UK as reported in the nationwide audit was common among patients with significant obesity. This is likely to be due to national guidelines that only support the use of GLP-1 receptor agonists in heavier patients.[2]
- Liraglutide showed effective glycaemic control across a range of overweight and obese body mass indices among noninsulin-treated patients with type 2 diabetes. The magnitude of HbA_{1c} improvement was comparable across baseline BMI categories.
- Absolute weight loss over 3 months of liraglutide treatment was significantly greater as baseline BMI increased.

References

1. Monami *et al. Exp Diabetes Res* 2012;2012:672658
2. NICE 2010. www.nice.org.uk/guidance/TA203

Acknowledgements

The ABCD nationwide GLP-1 receptor agonist audit programme has received grants from Eli Lilly & Co and Novo Nordisk A/S. This audit was independently initiated and performed by ABCD, and the authors remained independent in the analysis and writing of this report. The authors take full responsibility for the content of the poster but are grateful to Watermeadow Medical for editorial support in producing this poster (funded by Novo Nordisk).

Where EVERYONE Matters

Presented at the European Association for the Study of Diabetes, 1–5 October 2012, Berlin, Germany.